#5574

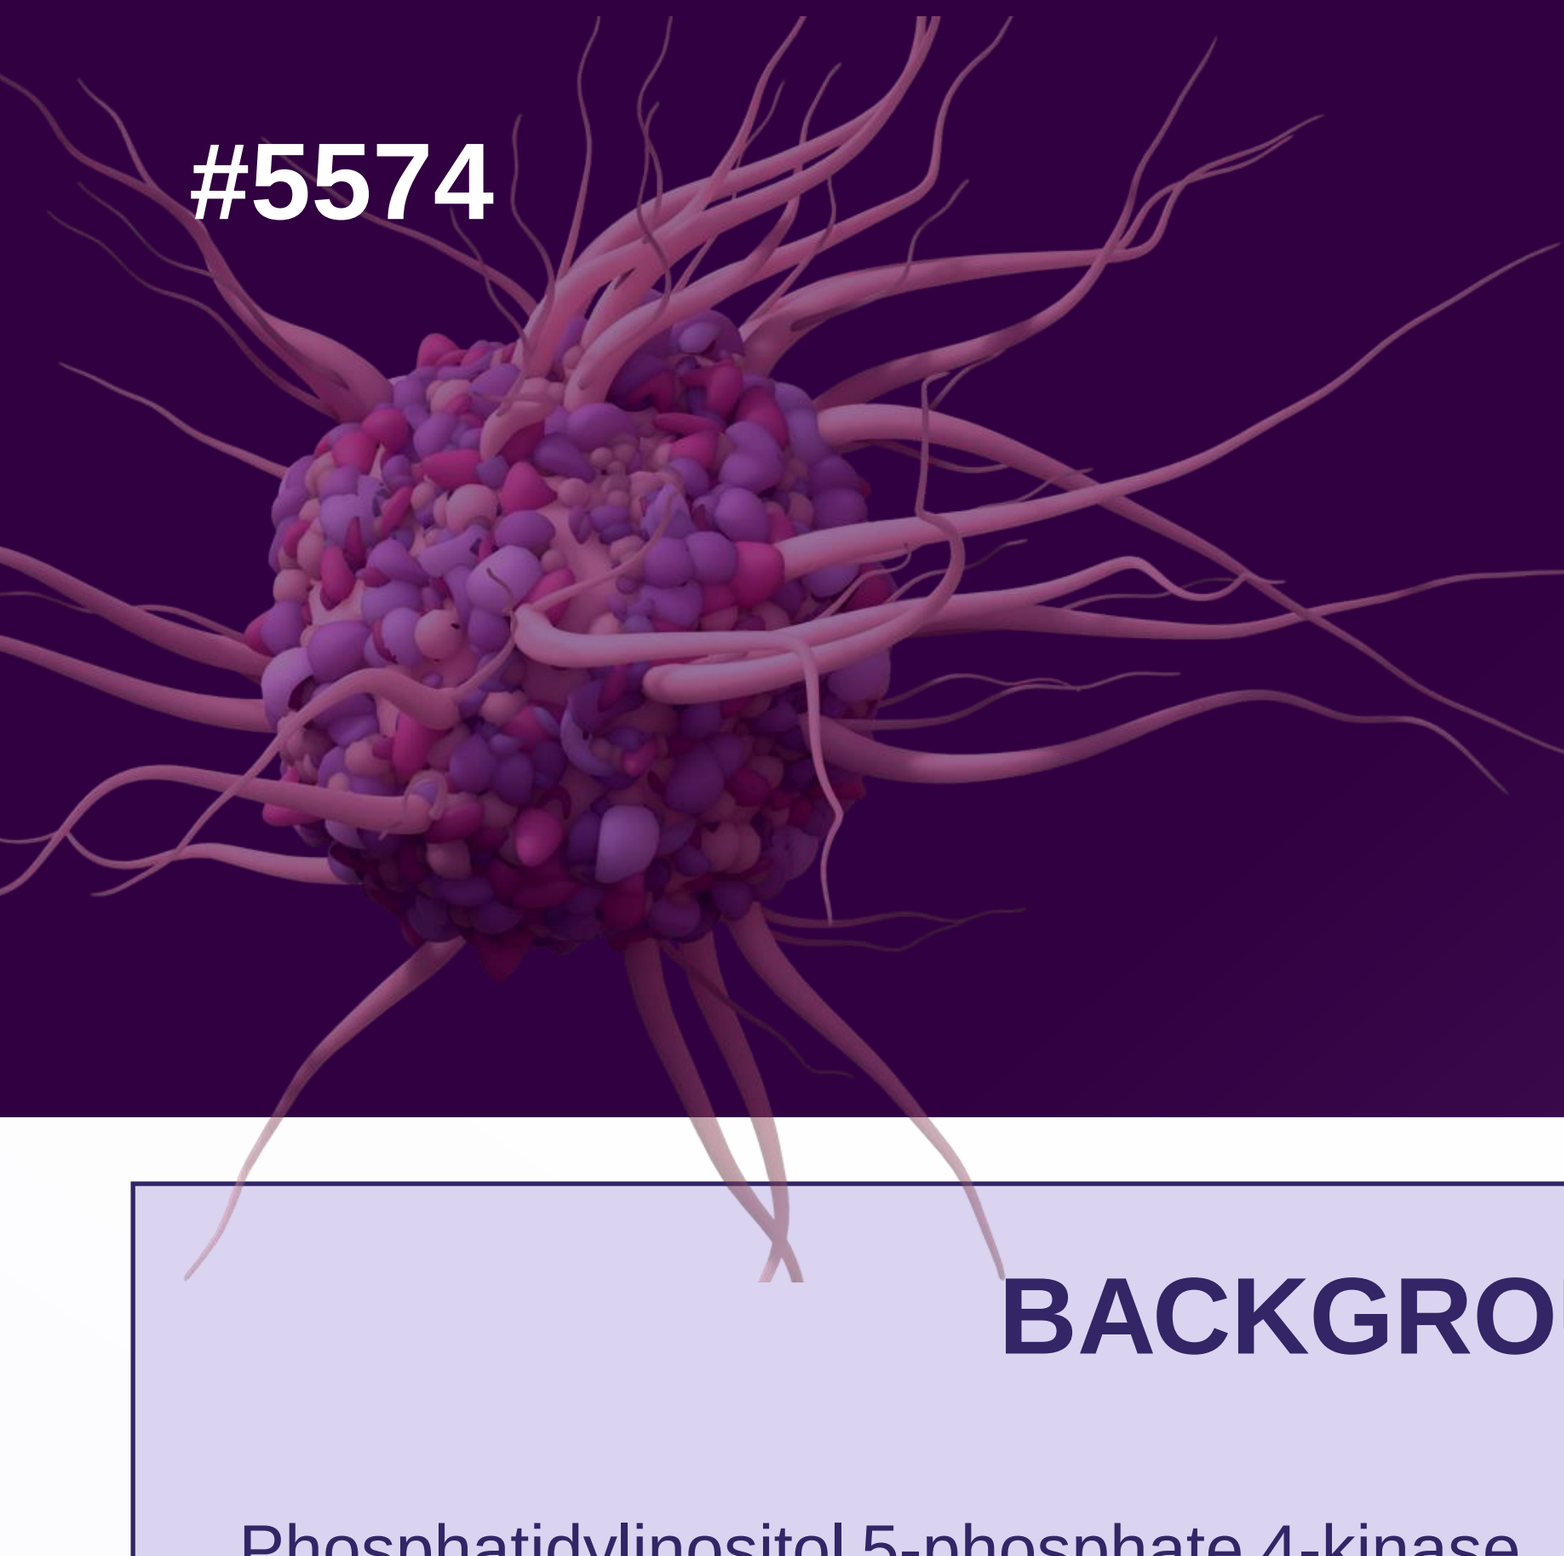

Degradation of PIP4K2C by novel bivalent functional degrader LRK-A induces tumor regression in CRC

Krista Goodman[1], Morgan O'Shea[1], Guosen Ye[1], Kristin Brown[1], Andrew Good[3], Melvyn Chow[1], Julie Arnold[1], Vincent Giacalone[1], Robert Prenovitz[1],Sebastian Hayes[1], Sydney Alnemy[1], Erica Dube[1], Oluwatimileyin Akinrinmade Adigun[1], Kirti Khardenavis[2], Alyssa Martin[2], Michael Perricone[2], **Eva d'Hennezel**[1], Emily Corse[1], Catherine Sabatos-Peyton[1].

[1] Larkspur Biosciences, Inc. Watertown, MA. [2] Xsphera Biosciences, Boston, MA. [3]CAnDiD Consulting, Wallingford, CT.

BACKGROUND

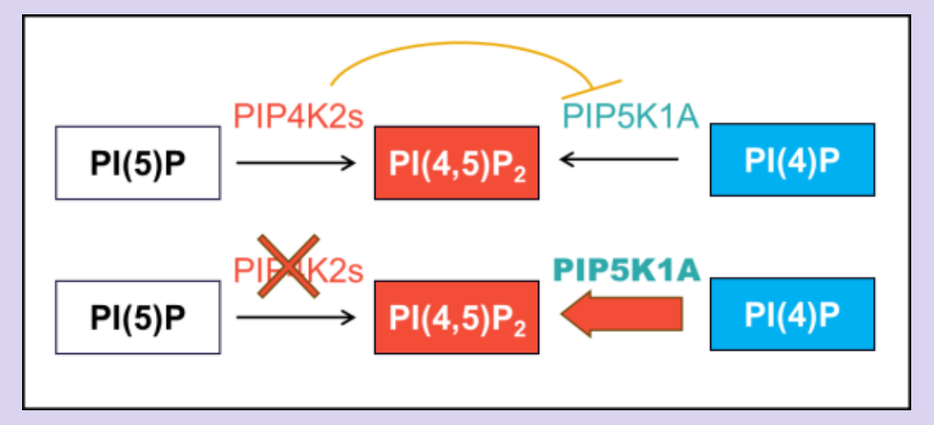
PI(5)P
PIP4K2s
PI(4,5)P2
PIP5K1A
PI(4)P

Phosphatidylinositol 5-phosphate 4-kinase, type II, gamma (PIP4K2C) is a lipid kinase with critical roles in vesicular trafficking, autophagy-dependent catabolism, and modulation of the immune system. Family members PIP4K2A and PIP4K2B are implicated in regulation of autophagy, cancer cell proliferation, and response to insulin, whereas PIP4K2C has a unique function in the immune response to cancer. Beyond the ability to convert PI(5)P to PI(4,5)P_2, PIP4K2 kinases regulate membrane localization and clustering of PI(4,5)P_2 and thus governs multiple aspects of membrane trafficking. These activities are independent of catalytic function[1]. The involvement of PIP4K2C in membrane lipid dynamics has the potential to broadly impact pro-tumor and immune-suppressive biology in cancer, including uptake of cancer cells by immune cells, and subsequently increase antigen processing, presentation, and T cell activation. Mice deficient in PIP4K2C develop immune cell infiltrates in tissues and increased proinflammatory cytokines in plasma with age[2], suggesting that modulation of PIP4K2C could enhance anti-tumor immunity in cancer patients.

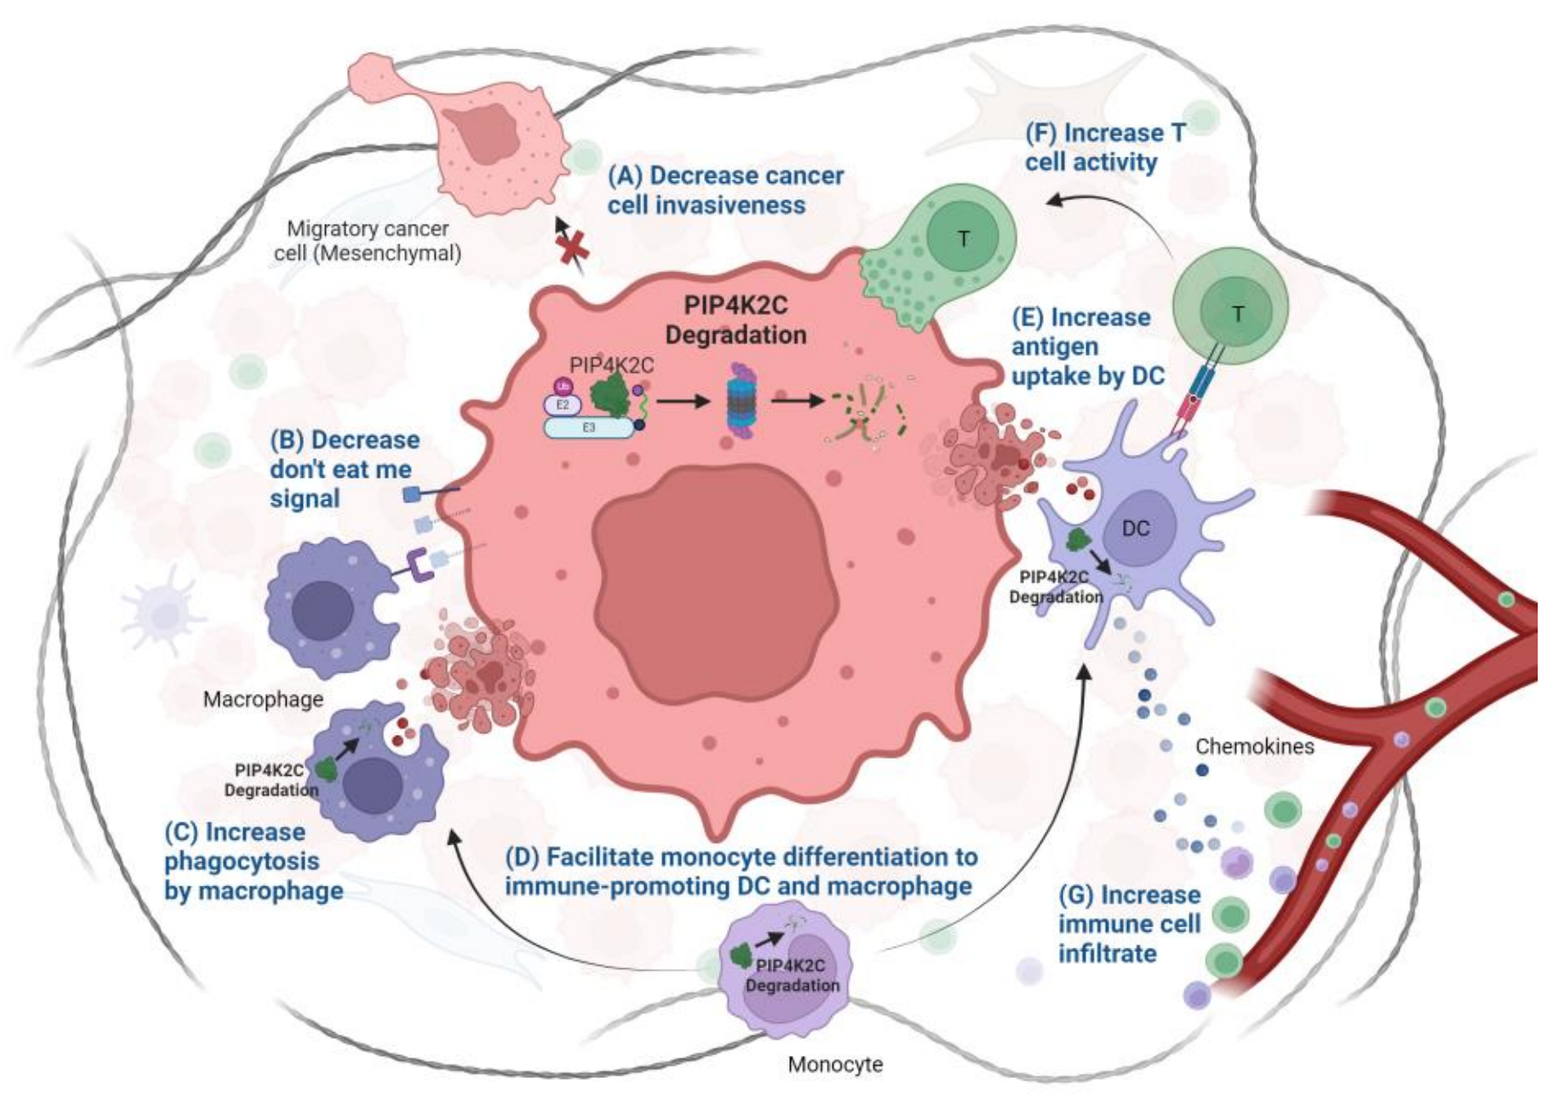
(A) Decrease cancer cell invasiveness
Migratory cancer cell (Mesenchymal)
(F) Increase T cell activity
PIP4K2C Degradation
(B) Decrease don't eat me signal
(E) Increase antigen uptake by DC
Macrophage
Chemokines
(C) Increase phagocytosis by macrophage
(D) Facilitate monocyte differentiation to immune-promoting DC and macrophage
(G) Increase immune cell infiltrate
Monocyte

Mechanistic model for targeting PIP4K2C in cancer. PIP4K2C abrogates the cancer's ability to grow and evade the immune system though multiple mechanisms, both from within the cancer cells and from the immune cells, resulting in a global rescue of anti-tumor immunity and reduced tumor growth.

1 CANCER-INTRINSIC IMPACT OF PIP4K2C DEFICIENCY

FIGURE 1a – Loss of PIP4K2C disrupts EMT transcriptional program.

Transcriptomic analyses reveal over 1200 genes (8.3% of total detected) differentially regulated by PIP4K2C knockout (KO) in SW837, a colorectal cancer (CRC) cell line. Pathways critical for cancer progression are modulated, including increased interferon response (e. g. *IFITM1, OAS1, IRF1*), and reduced epithelial-to-mesenchymal transition (EMT) (e. g. *VIM, CXCL1, SPOCK1, VEGFC*) and DAG/IP3 signaling (e. g. *ADCY, PLCG, PRKCA/B*) suggesting that targeting PIP4K2C in cancer patients may have pleiotropic impacts.

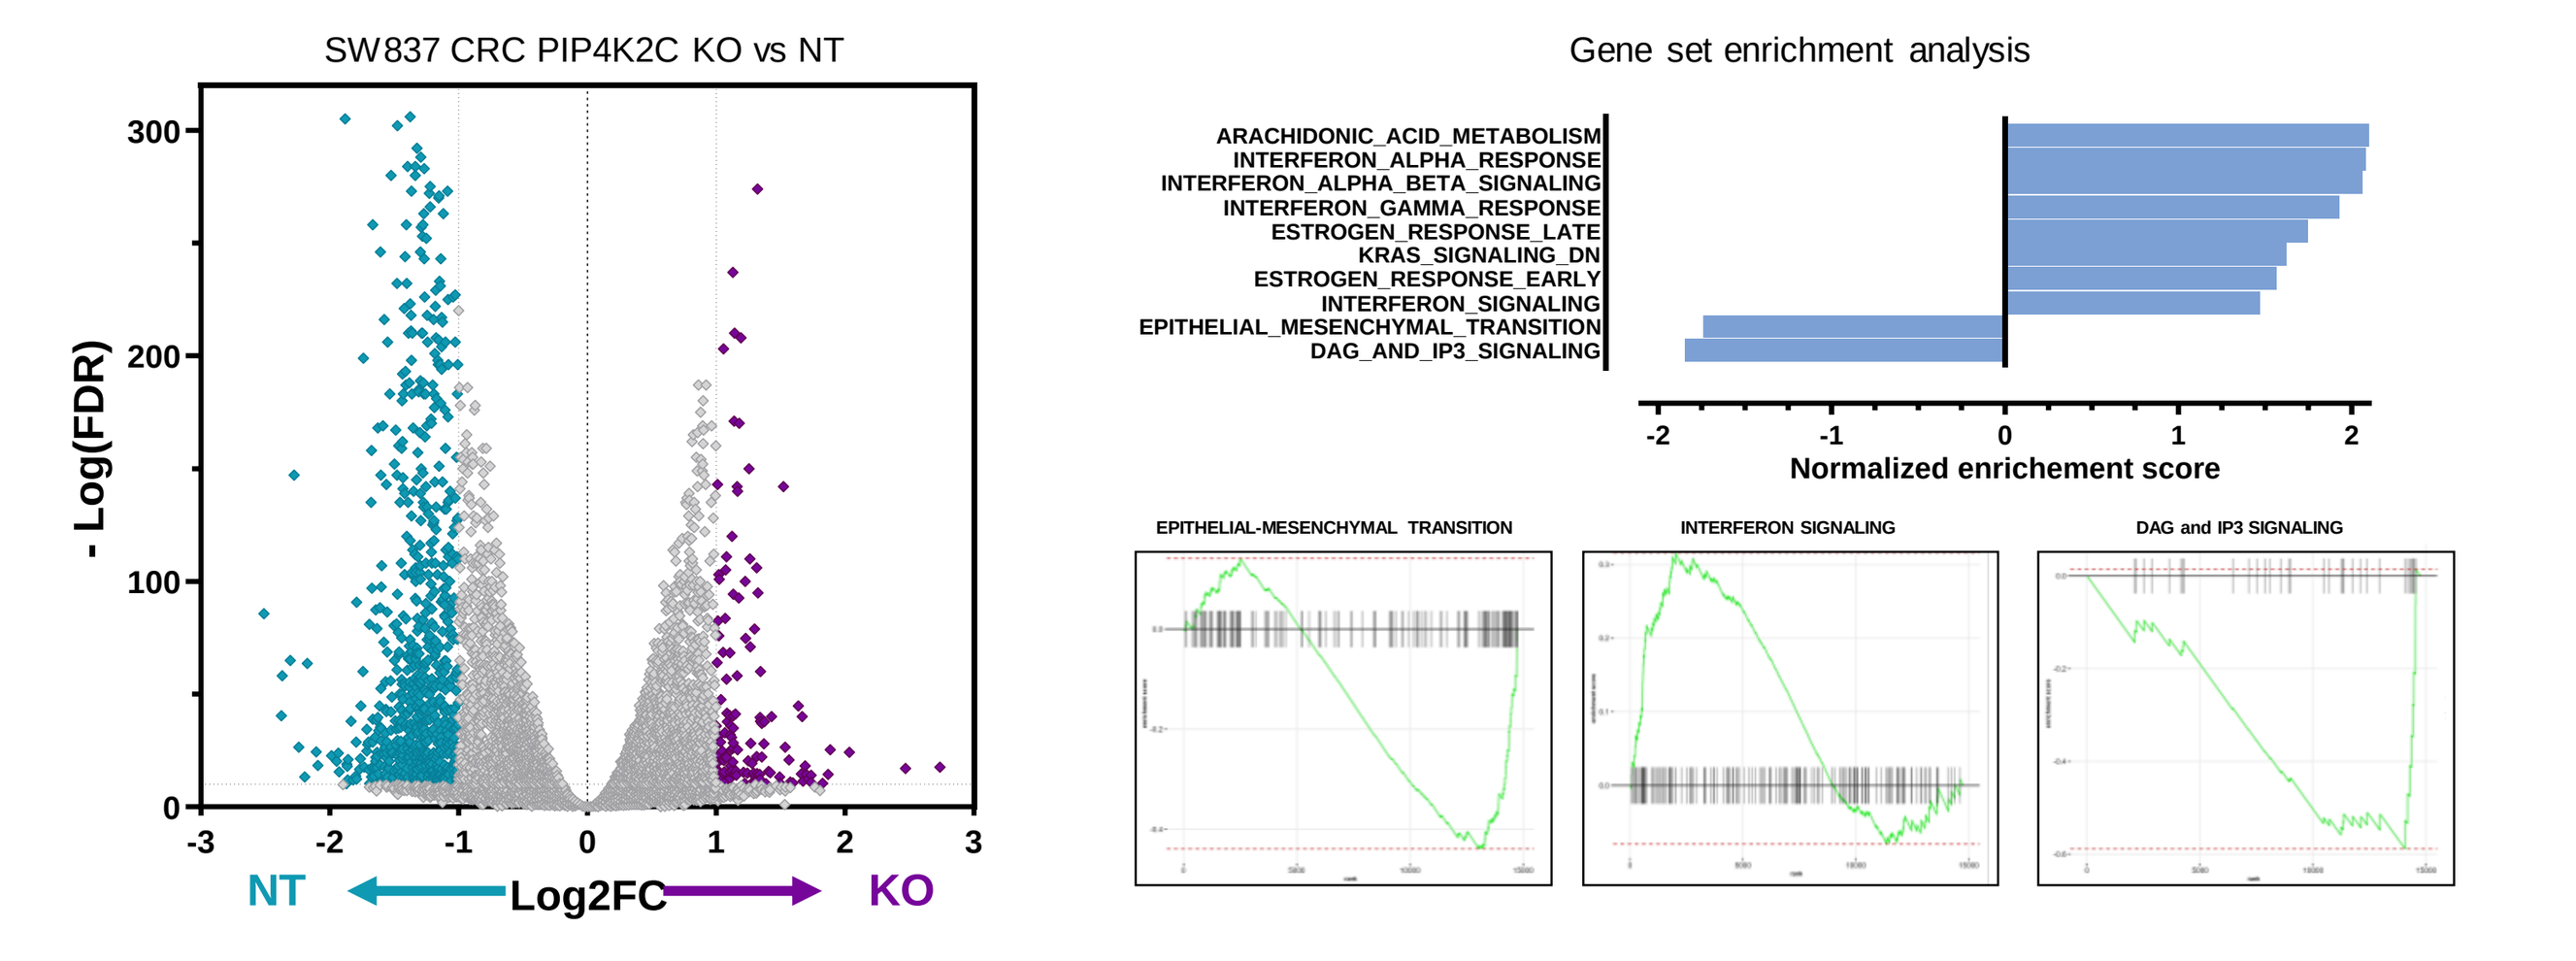
SW837 CRC PIP4K2C KO vs NT
- Log(FDR)
NT
Log2FC
KO
Gene set enrichment analysis
ARACHIDONIC_ACID_METABOLISM
INTERFERON_ALPHA_RESPONSE
INTERFERON_ALPHA_BETA_SIGNALING
INTERFERON_GAMMA_RESPONSE
ESTROGEN_RESPONSE_LATE
KRAS_SIGNALING_DN
ESTROGEN_RESPONSE_EARLY
INTERFERON_SIGNALING
EPITHELIAL_MESENCHYMAL_TRANSITION
DAG_AND_IP3_SIGNALING
Normalized enrichement score
EPITHELIAL-MESENCHYMAL TRANSITION
INTERFERON SIGNALING
DAG and IP3 SIGNALING

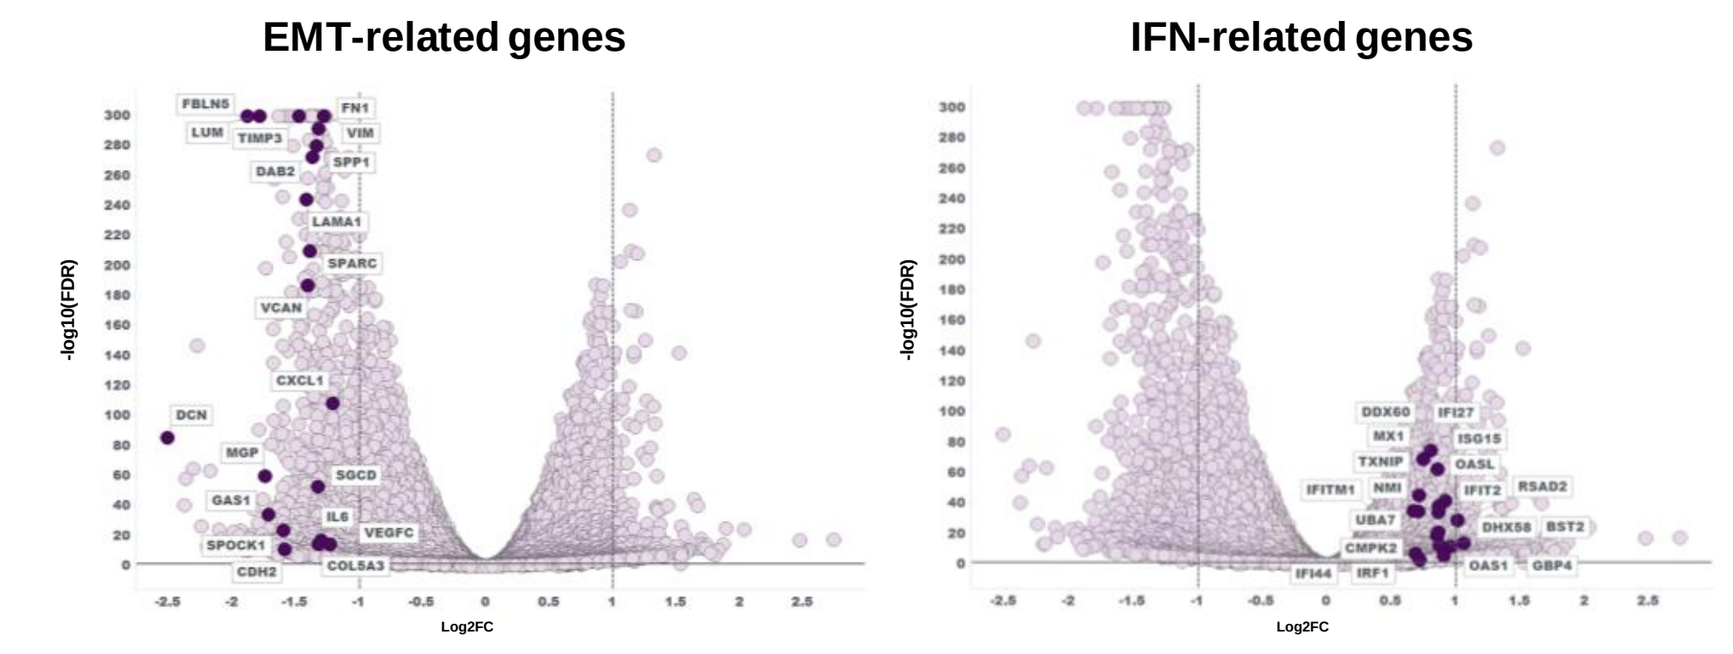
EMT-related genes
IFN-related genes

PIP4K2C KO (CRISPR/RNP nucleofection) or control (non-targeting sgRNA) SW837 CRC cells were analyzed by RNAseq (Illumina, Medgenome). 1222 of 14667 detected genes are modulated by PIP4K2C deficiency (left, top). Pathway enrichment analysis was performed using REACTOME, HARMONY and KEGG pathway databases (right, top). Key genes implicated in top modulated pathways are indicated (left, bottom).

FIGURE 1b – PIP4K2C deficiency alters cancer secretome to reduce EMT and enhance anti-tumor immunity

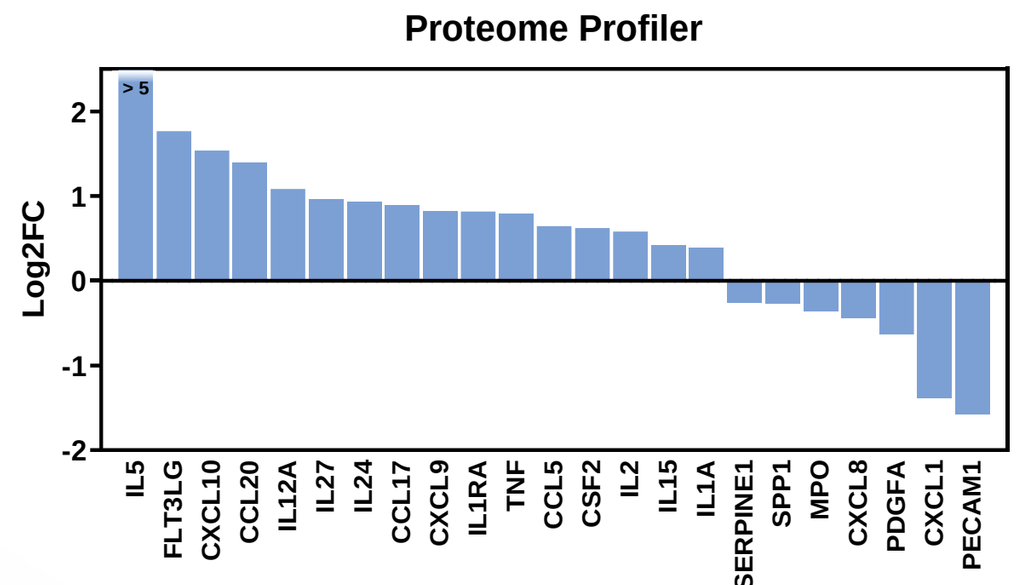
Proteome Profiler
Log2FC
IL5
FLT3LG
CXCL10
CCL20
IL12A
IL27
IL24
CCL17
CXCL9
IL1RA
TNF
CCL5
CSF2
IL2
IL15
IL1A
SERPINE1
SPP1
MPO
CXCL8
PDGFA
CXCL1
PECAM1

In accordance with transcriptionally modulated pathways, Proteome profiler analysis of culture supernatants reveals increased pro-inflammatory cytokines and chemokines (e.g. FLT3L, CXCL9, CXCL10, IL5) and reduced pro-tumor invasiveness factors (CXCL1, CD31) in PIP4K2C KO cancer cells.

Culture supernatant from PIP4K2C KO (CRISPR/RNP nucleofection) or control (non-targeting sgRNA) SW837 CRC cells were analyzed by semi-quantitative Proteome Profiler (R&D Systems).

2 COOPERATIVE CANCER AND IMMUNE REGULATION

FIGURE 2a – Loss of PIP4K2C in either cancer or myeloid cells enhances phagocytosis

Dendritic cells more actively phagocytose dying PIP4K2C KO tumor cells compared to non-targeted (NT) control cancer cells, highlighting the effect of PIP4K2C on cancer immunogenicity. The degradation of PIP4K2C in human dendritic cells and macrophages enhanced their phagocytosis of bioparticles, emphasizing the intrinsic immunological benefits of PIP4K2C targeting in myeloid cells. Importantly, employing a control that binds but does not degrade PIP4K2C underscores the advantage of utilizing degrader strategies to target PIP4K2C effectively.

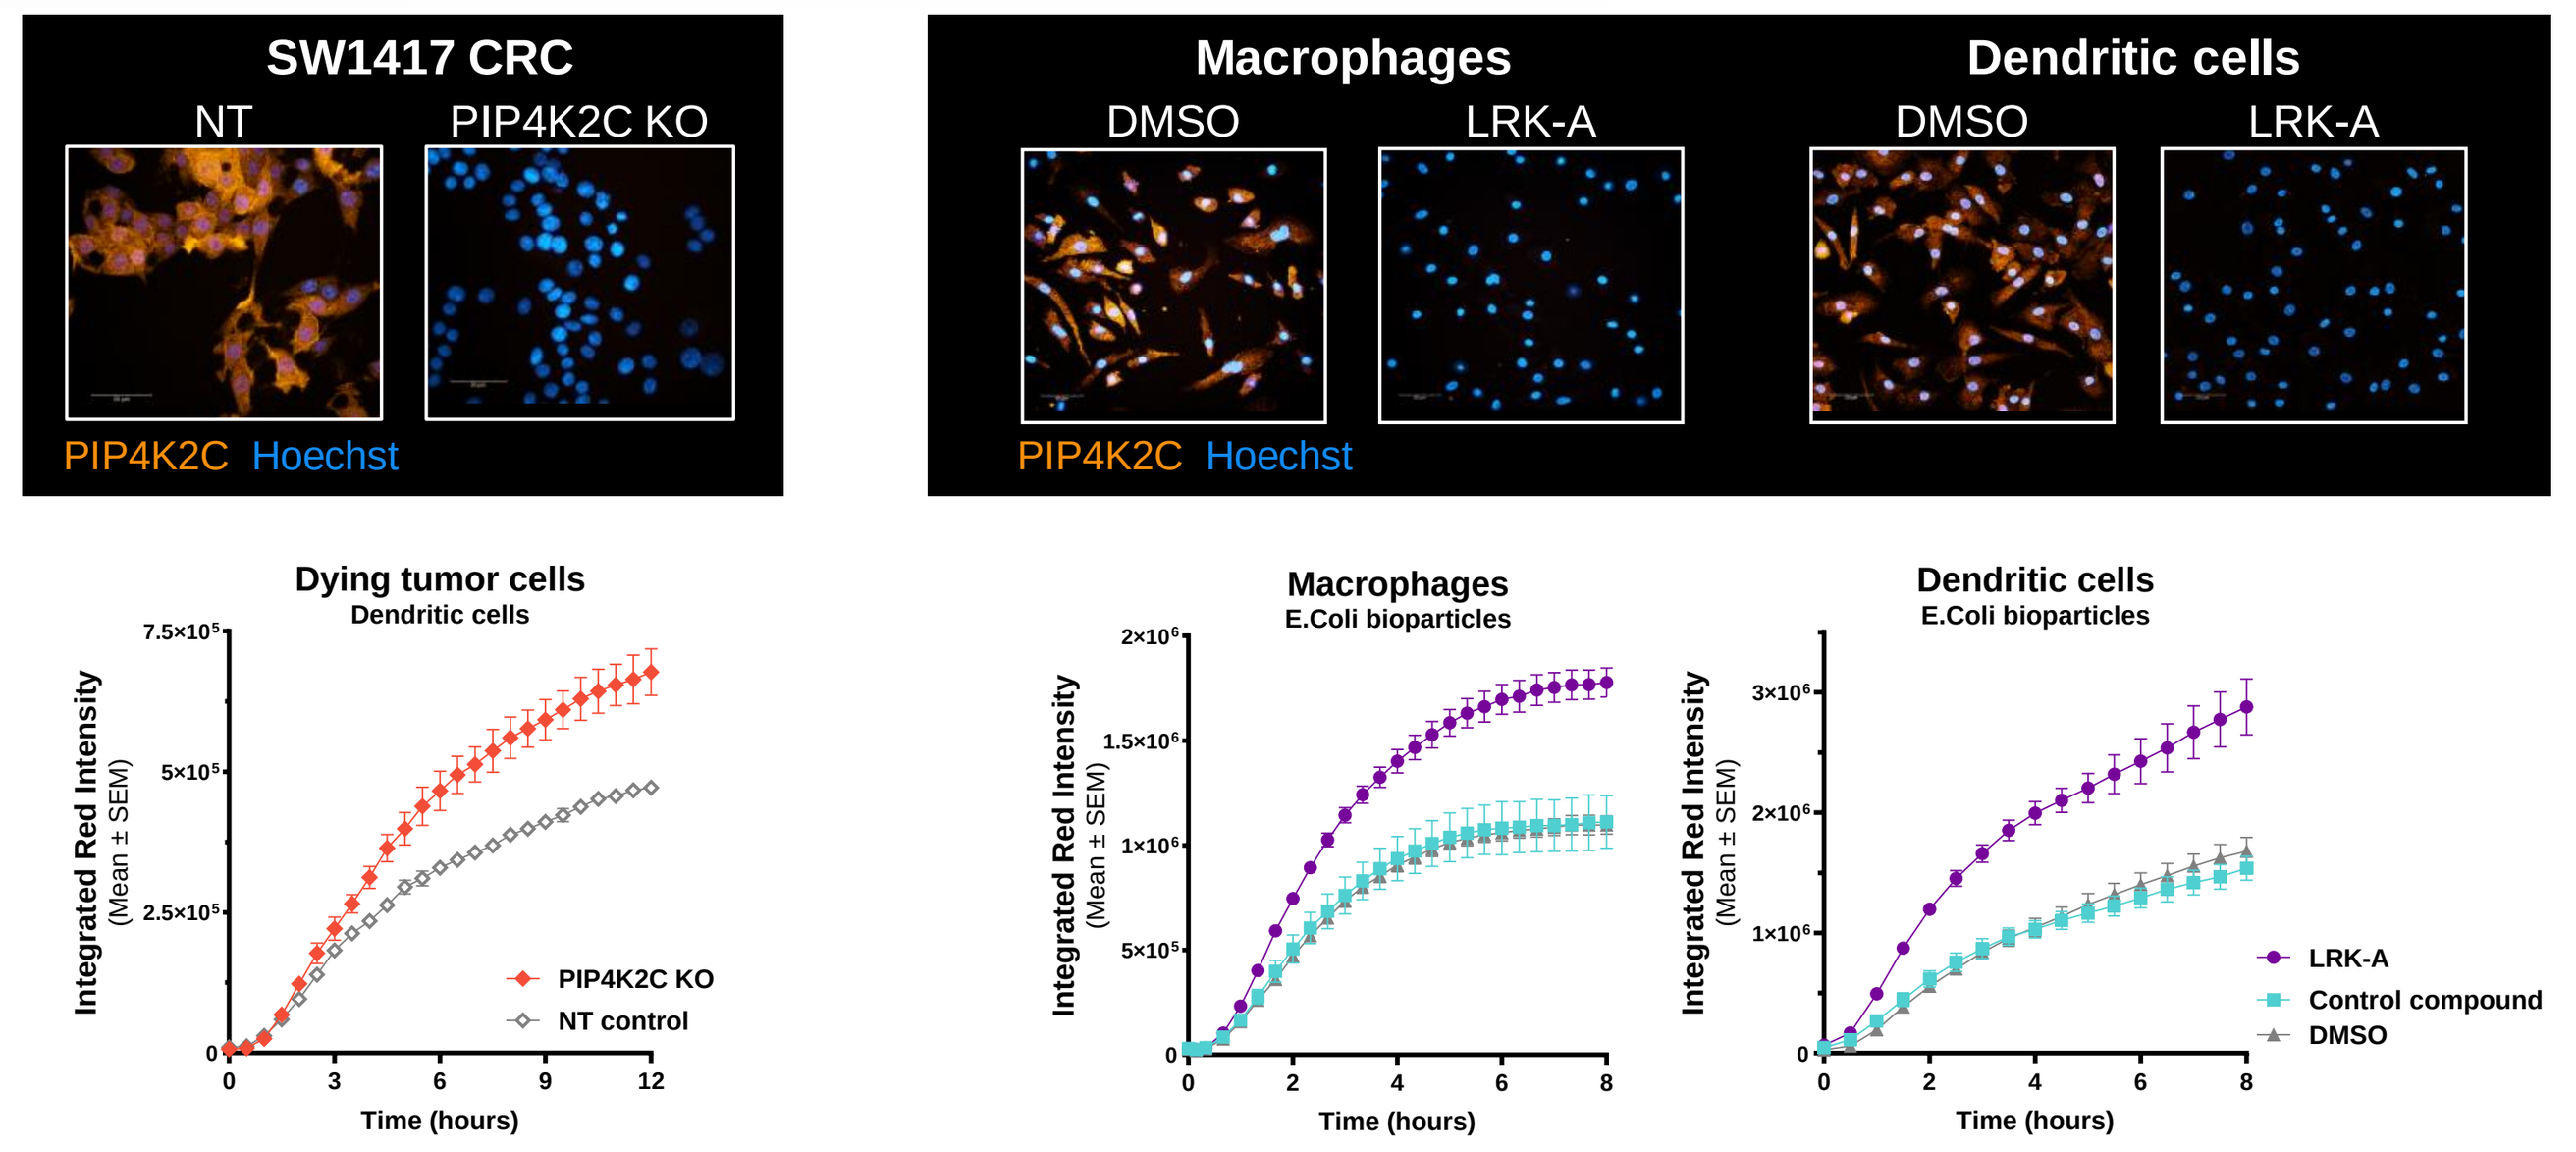
SW1417 CRC
NT
PIP4K2C KO
PIP4K2C Hoechst
Macrophages
DMSO
LRK-A
Dendritic cells
DMSO
LRK-A
PIP4K2C Hoechst
Dying tumor cells
Dendritic cells
Integrated Red Intensity (Mean ± SEM)
PIP4K2C KO
NT control
Time (hours)
Macrophages
E.Coli bioparticles
Dendritic cells
E.Coli bioparticles
LRK-A
Control compound
DMSO

(left) Uptake of dying (H_2O_2 2 nM, 4 hrs) pHrodo red-labelled SW1417 tumor cells either PIP4K2C KO (CRISPR) or non-targeting control, by monocyte-derived dendritic cells. Uptake monitored by live cell imaging (Incucyte). (right) Primary human monocytes derived in vitro into macrophages or immature dendritic cells in the presence of DMSO, the PIP4K2C degrader LRK-A or a non-degrading control (50 nM); phagocytosis of bioparticles labeled with pHrodo red is shown. Top: Loss of PIP4K2C by CRIPSR (left) or by degradation (right) was verified by immunofluorescence (representative images, 40X).

FIGURE 2b – Lack of PIP4K2C increases PI(4,5)P_2 during phagocytosis

Consistent with the current understanding of PIP4K2C's molecular mode of action, when dendritic cells are differentiated in the absence of PIP4K2C, levels of PI(4,5)P_2 actively increase during phagocytosis compared to cells derived with DMSO. This elevation in PI(4,5)P_2 is aligned with increased phagocytic activity, suggesting that the reported ability of PI(4,5)P_2 to modulate plasma membrane dynamics is involved in the increased phagocytic ability observed in the absence of PIP4K2C.

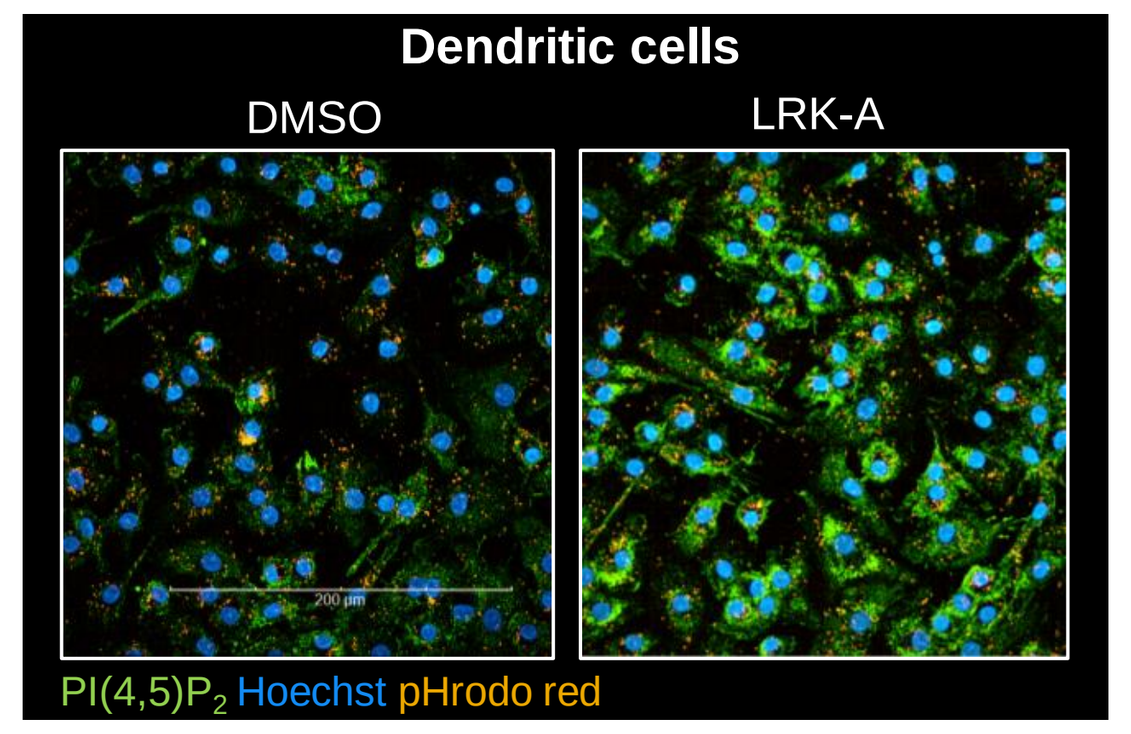
Dendritic cells
DMSO
LRK-A
PI(4,5)P2 Hoechst pHrodo red

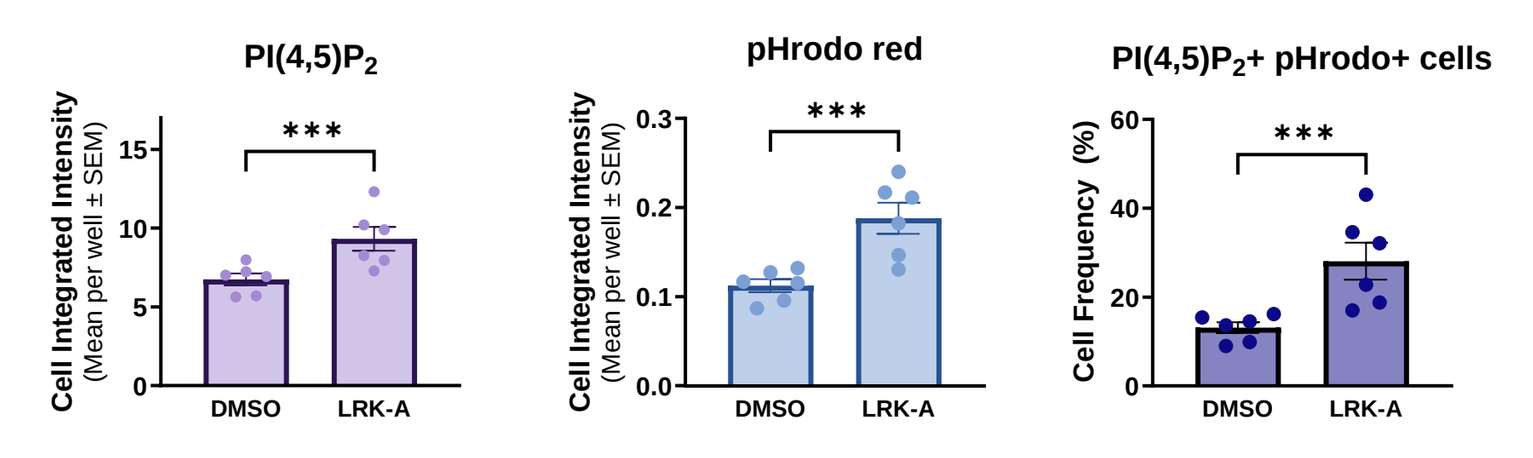
PI(4,5)P2
pHrodo red
PI(4,5)P2+ pHrodo+ cells
Cell Integrated Intensity (Mean per well ± SEM)
Cell Frequency (%)
DMSO
LRK-A

Primary human monocytes differentiated into immature dendritic cells in the presence of DMSO or the PIP4K2C degrader LRK-A (50 nM) were fed pHrodo red-labelled bioparticles for phagocytosis. After 30 min of incubation, engulfment of bioparticles and PI(4,5)P_2 levels were measured by immunofluorescence (Operetta). (left) Representative immunofluorescence staining (40X, confocal) showing dendritic cells probed for PI(4,5)P_2 and pHrodo. (right) Quantification of PI(4,5)P_2 and pHrodo red in individual cells, averaged per well (n=6 wells). *** t-test p<0.005

FIGURE 2c - PIP4K2C deficiency acts cooperatively in tumors and dendritic cells to enhance tumor immunogenicity

Absence of PIP4K2C in dendritic cells leads to an increased uptake of dying tumor-derived material, adding to the enhanced engulfment of PIP4K2C-deficient tumor cells by immune cells. This additive impact maximizes tumor cell clearance, antigen presentation and immune stimulation when targeting PIP4K2C in both tumor and immune cells.

This mechanism adds to the cancer-specific impacts identified in transcriptional studies (Figure 1), revealing a broad spectrum of effects from targeting PIP4K2C: tumor-intrinsic, immune-driven by tumors, and directly related to the immune system, highlighting multifaceted, potent impact of targeting PIP4K2C in cancer therapy.

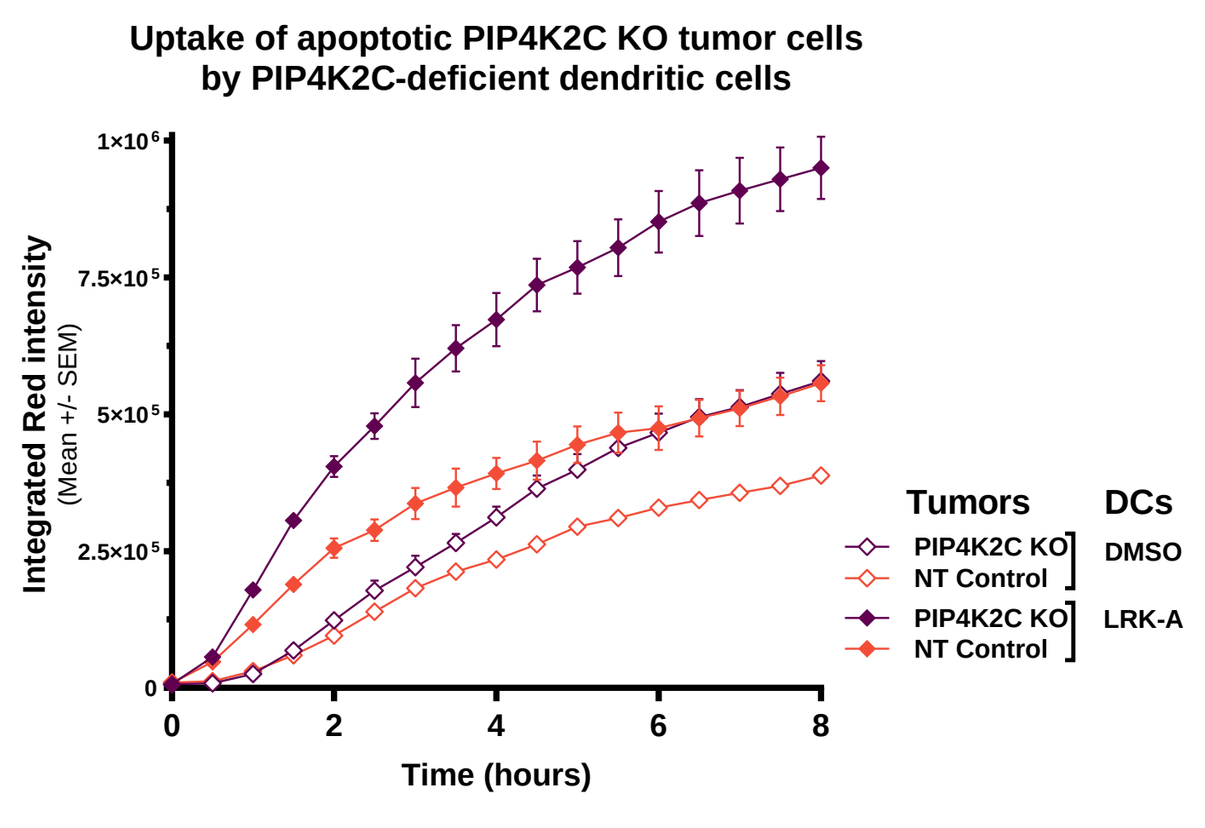
Uptake of apoptotic PIP4K2C KO tumor cells by PIP4K2C-deficient dendritic cells
Integrated Red intensity (Mean +/- SEM)
Time (hours)
Tumors
DCs
PIP4K2C KO
NT Control
DMSO
PIP4K2C KO
NT Control
LRK-A

Primary human monocytes derived in vitro into immature dendritic cells in the presence of LRK-A (50 nM) or DMSO. Phagocytosis of SW1417 CRC cells stably KO for PIP4K2C or NT control and labelled with pHrodo red dye, after 4 hr H_2O_2 (2 mM).

3 LRK-A : FIRST-IN-CLASS DEVELOPMENT CANDIDATE

LRK-A is a first-in-class bivalent cereblon (CRBN)-based degrader of PIP4K2C which drives rapid and deep degradation in all species evaluated. LRK-A is highly selective against other kinases and degradation off-targets as established by whole-cell proteomics.

Compound Profile

PIP4K2C DC_{50} (D_{max}, MOLT4)	4.3 nM, 85% (6h) 4.5 nM, 84% (24h)
PIP4K2C PBMC DC_{50} (D_{max})	0.3 nM, 78% (human, 6h) 5.1 nM, 84% (mouse, 6h)
F_u (h, m, r, d, c)	4%, 3%, 8%, 6%, 8%
hERG manual patch clamp	11% inhibition at 30 uM
Degradation selectivity (MOLT4, PBMC)	**Fully selective for PIP4K2C**
Kinase selectivity	Selective vs >100 kinases
Cereblon off-targets	No degradation of SALL-4 or GSPT-1
Degradation in preclinical species	Confirmed in mouse, rat, dog
Human dose projection	**< 10 mg**

Whole-cell proteomics

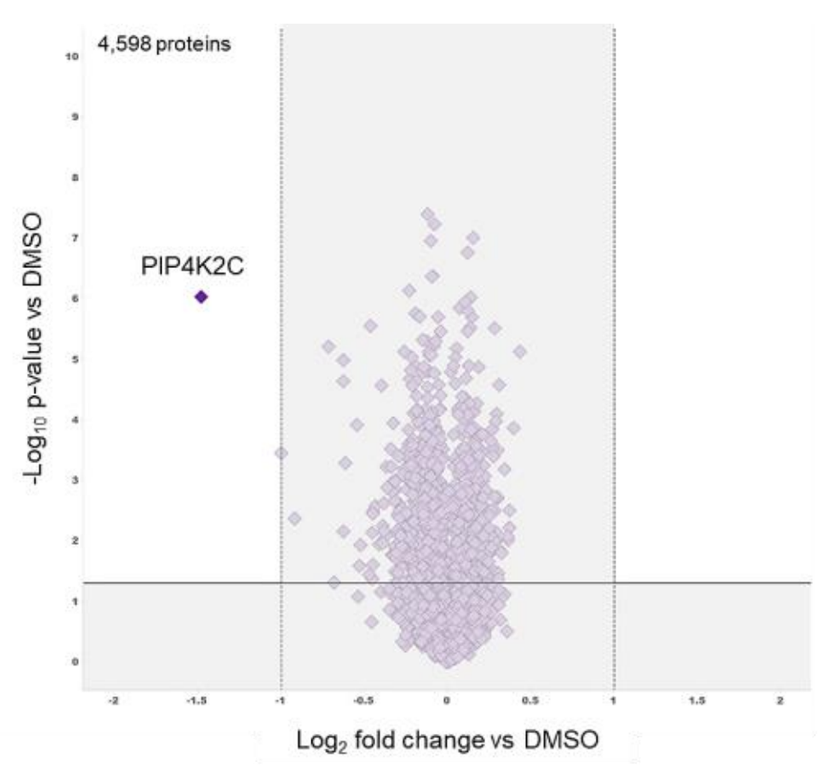
4,598 proteins
PIP4K2C
$-Log_{10}$ p-value vs DMSO
Log_2 fold change vs DMSO

MOLT4 cells were incubated with LRK-A (100 nM) for 6 hrs and analyzed for protein degradation by TMT-LC-MS. LRK-A specifically degrades PIP4K2C alone amongst 4598 proteins detected. Similar results obtained in PBMC from 3 independent donors (not shown).

FIGURE 3a – LRK-A is a potent degrader of endogenous PIP4K2C across species

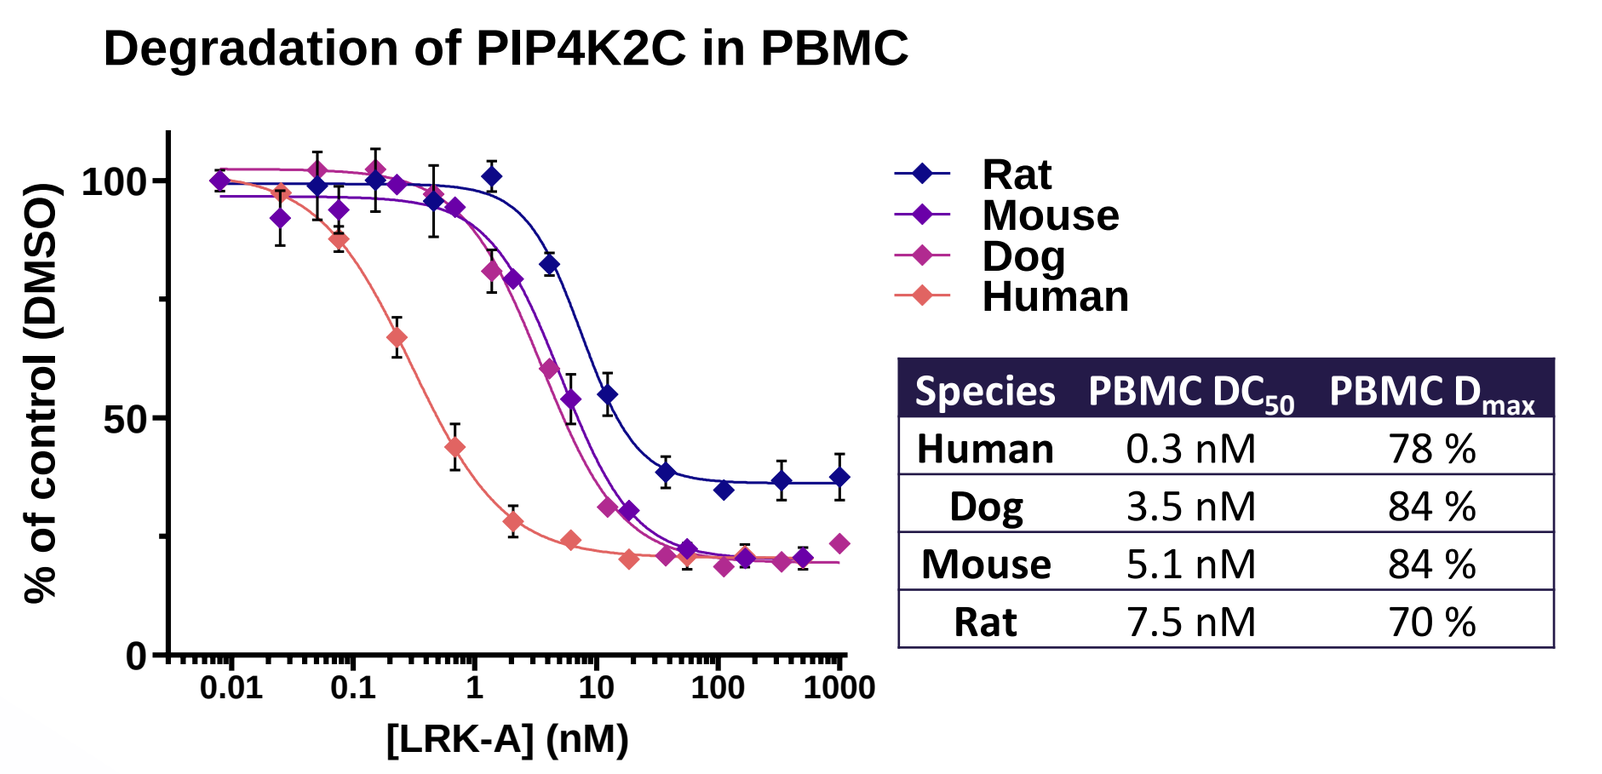
Degradation of PIP4K2C in PBMC
% of control (DMSO)
[LRK-A] (nM)
Rat
Mouse
Dog
Human

Species	PBMC DC_{50}	PBMC D_{max}
Human	0.3 nM	78 %
Dog	3.5 nM	84 %
Mouse	5.1 nM	84 %
Rat	7.5 nM	70 %

Primary PBMC from indicated species were incubated with a range of doses of LRK-A for 6 hrs. PIP4K2C was detected by intracellular flow cytometry.

LRK-A degraded PIP4K2C rapidly and profoundly in PBMCs from multiple species. Differences in potency between species are observed which are in line with the current understanding of species-specific variations in CRBN sequence and subsequent function for targeted protein degradation (mouse ~17-fold < human).

FIGURE 3b – Single dose of LRK-A drives profound and sustained loss of PIP4K2C in vivo

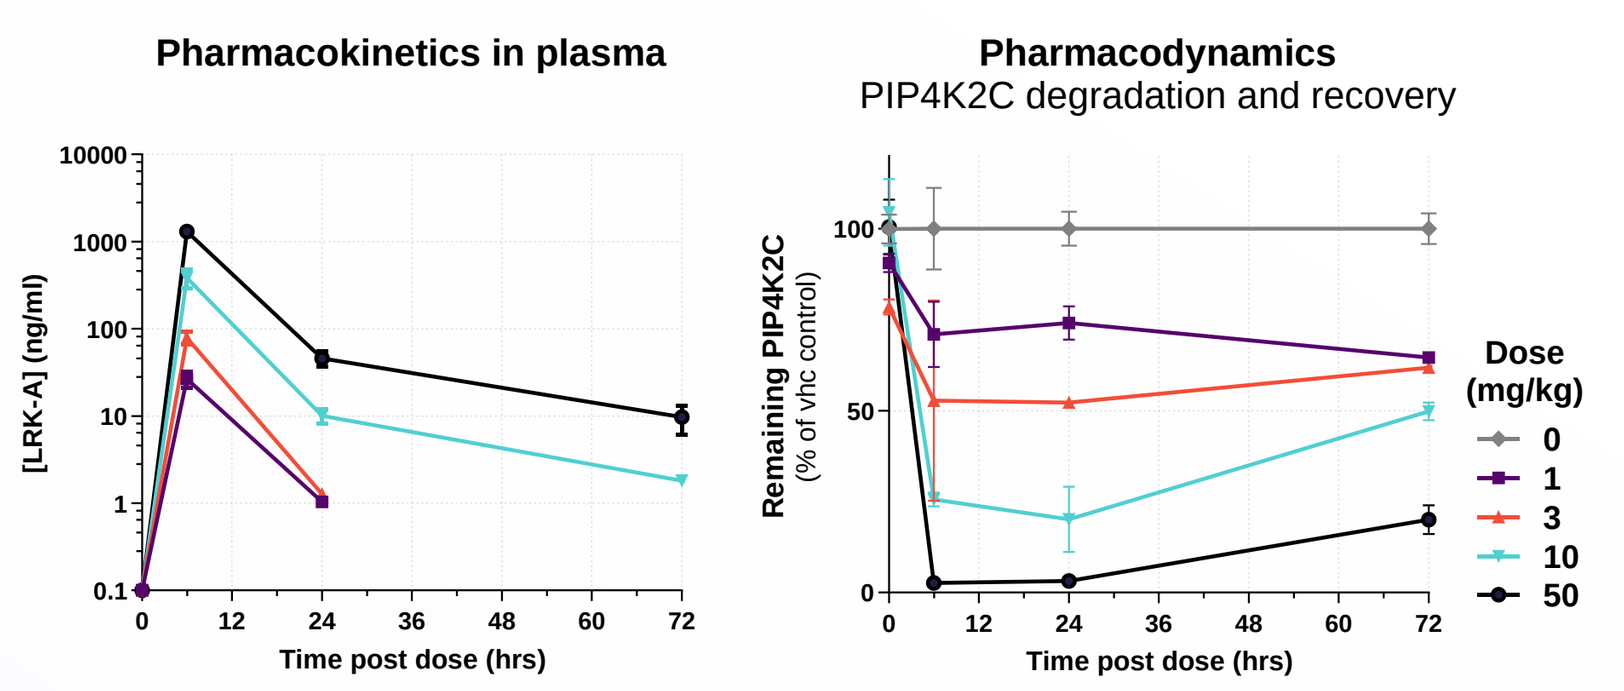
Pharmacokinetics in plasma
[LRK-A] (ng/ml)
Time post dose (hrs)
Pharmacodynamics
PIP4K2C degradation and recovery
Remaining PIP4K2C (% of vhc control)
Dose (mg/kg)
0
1
3
10
50

Mice received a single dose of LRK-A i.p. at different concentrations as indicated. Plasma levels of LRK-A were measured by mass spectrometry and cellular levels of PIP4K2C were measured by intracellular flow cytometry at the indicated timepoints. Data is represented from n=3 animals at each time point. Similar results obtained in spleen (data not shown).

A single dose of LRK-A is sufficient to ablate PIP4K2C in peripheral PBMC in mice. Degradation is rapid (max $\leq$ 6hrs) and sustained for up to 72hrs post-dose at every dose tested, suggesting feasibility of an intermittent dosing regimen. Pharmacokinetics indicate a dose-proportionate increase in exposure and suggest that a low exposure over a short time is sufficient to drive a strong and sustained degradation in vivo.

FIGURE 3c – LRK-A is efficacious at low dose in a mouse model of colon carcinoma

Administration of LRK-A (3 mg/kg i.p. Q2D) to mice with established tumors significantly hindered tumor growth and induced complete regression in the MC38-OVA syngeneic CRC model, leading to improved survival. In line with PK/PD data reported above, the evaluation of PIP4K2C levels in PBMCs demonstrates that the observed anti-tumor efficacy is associated with approximately 50% target degradation in blood.

Study Design:
- MC38-OVA CRC syngeneic model (subcutaneous)
- Q2D i.p. dosing starting on day 20 (established tumors > 100 mm³)
- Tumor volumes measured by caliper every other day
- PBMC collection for PD in life on day 9 of treatment (steady state)

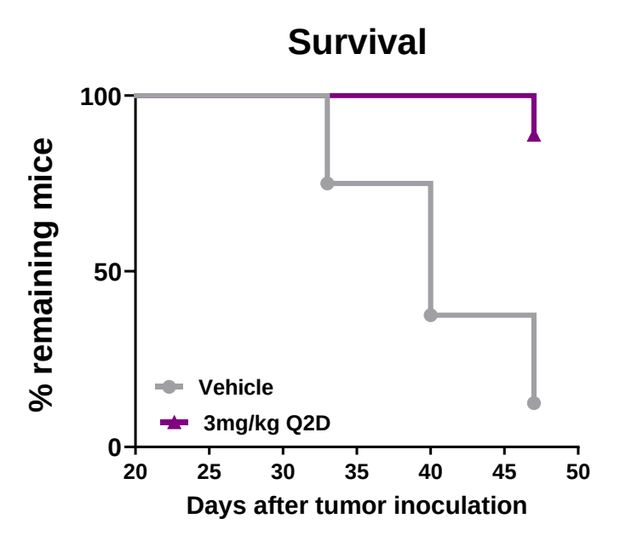
Survival
% remaining mice
Vehicle
3mg/kg Q2D
Days after tumor inoculation

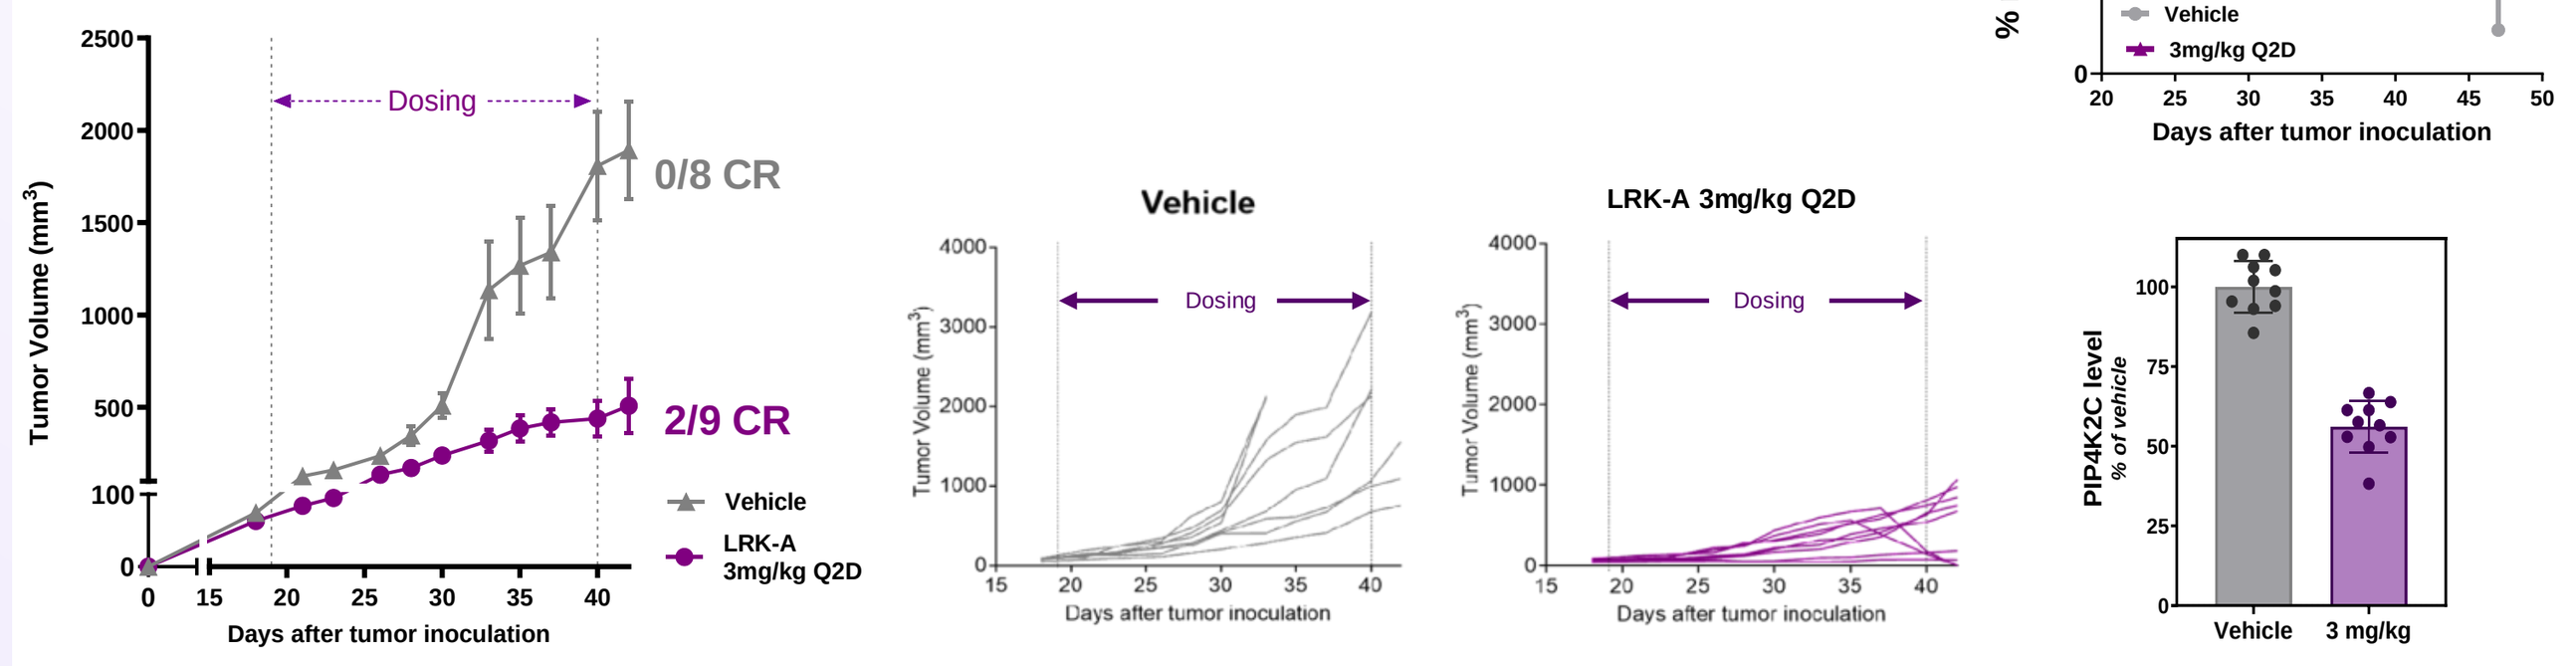
Dosing
0/8 CR
2/9 CR
Tumor Volume (mm³)
Vehicle
LRK-A 3mg/kg Q2D
Days after tumor inoculation
Vehicle
LRK-A 3mg/kg Q2D
PIP4K2C level % of vehicle
Vehicle
3 mg/kg

Mice bearing established tumors of 80-100 mm³ were randomized into groups one day prior to initiation of dosing, on day 19 after tumor implantation. LRK-A was administered Q2D for 21 days and tumor growth was monitored by caliper. Average tumor sizes (left) and individual growth curves (center) are shown. (Upper right) Kaplan-Meier survival curve. (Lower right) Degradation confirmed in PBMC by flow cytometry on day 15 of dosing, 24 hrs post dose.

4 HUMAN EFFICACY PoC IN CRC PATIENT SAMPLES

Multi-channel microfluidics platform
Primary patient-derived organoid spheres (resections)
Multiplexing
- Baseline flow cytometry immunophenotyping
- Cytotoxic response
- Transcriptional readout post-treatment (Nanostring IO panel)

FIGURE 4a – LRK-A drives cytotoxicity in human CRC patient ex vivo spheroids

Treatment of primary, treatment-naïve CRC resection-derived spheroids with LRK-A for up to 5 days led to profound degradation of PIP4K2C, and significant cytotoxicity in 5/7 patients tested to date. Importantly, LRK-A does not have direct cytolytic effects on cancer cells, suggesting these responses are immune-mediated. These preliminary data compare favorably to the Cetuximab benchmark of the platform, which captures a clinically-relevant efficacy of 35% (6/16 responders) in *KRAS*^{WT} CRC. Together, these results demonstrate that targeting PIP4K2C with the degrader LRK-A has the potential to improve the outcomes in CRC patients.

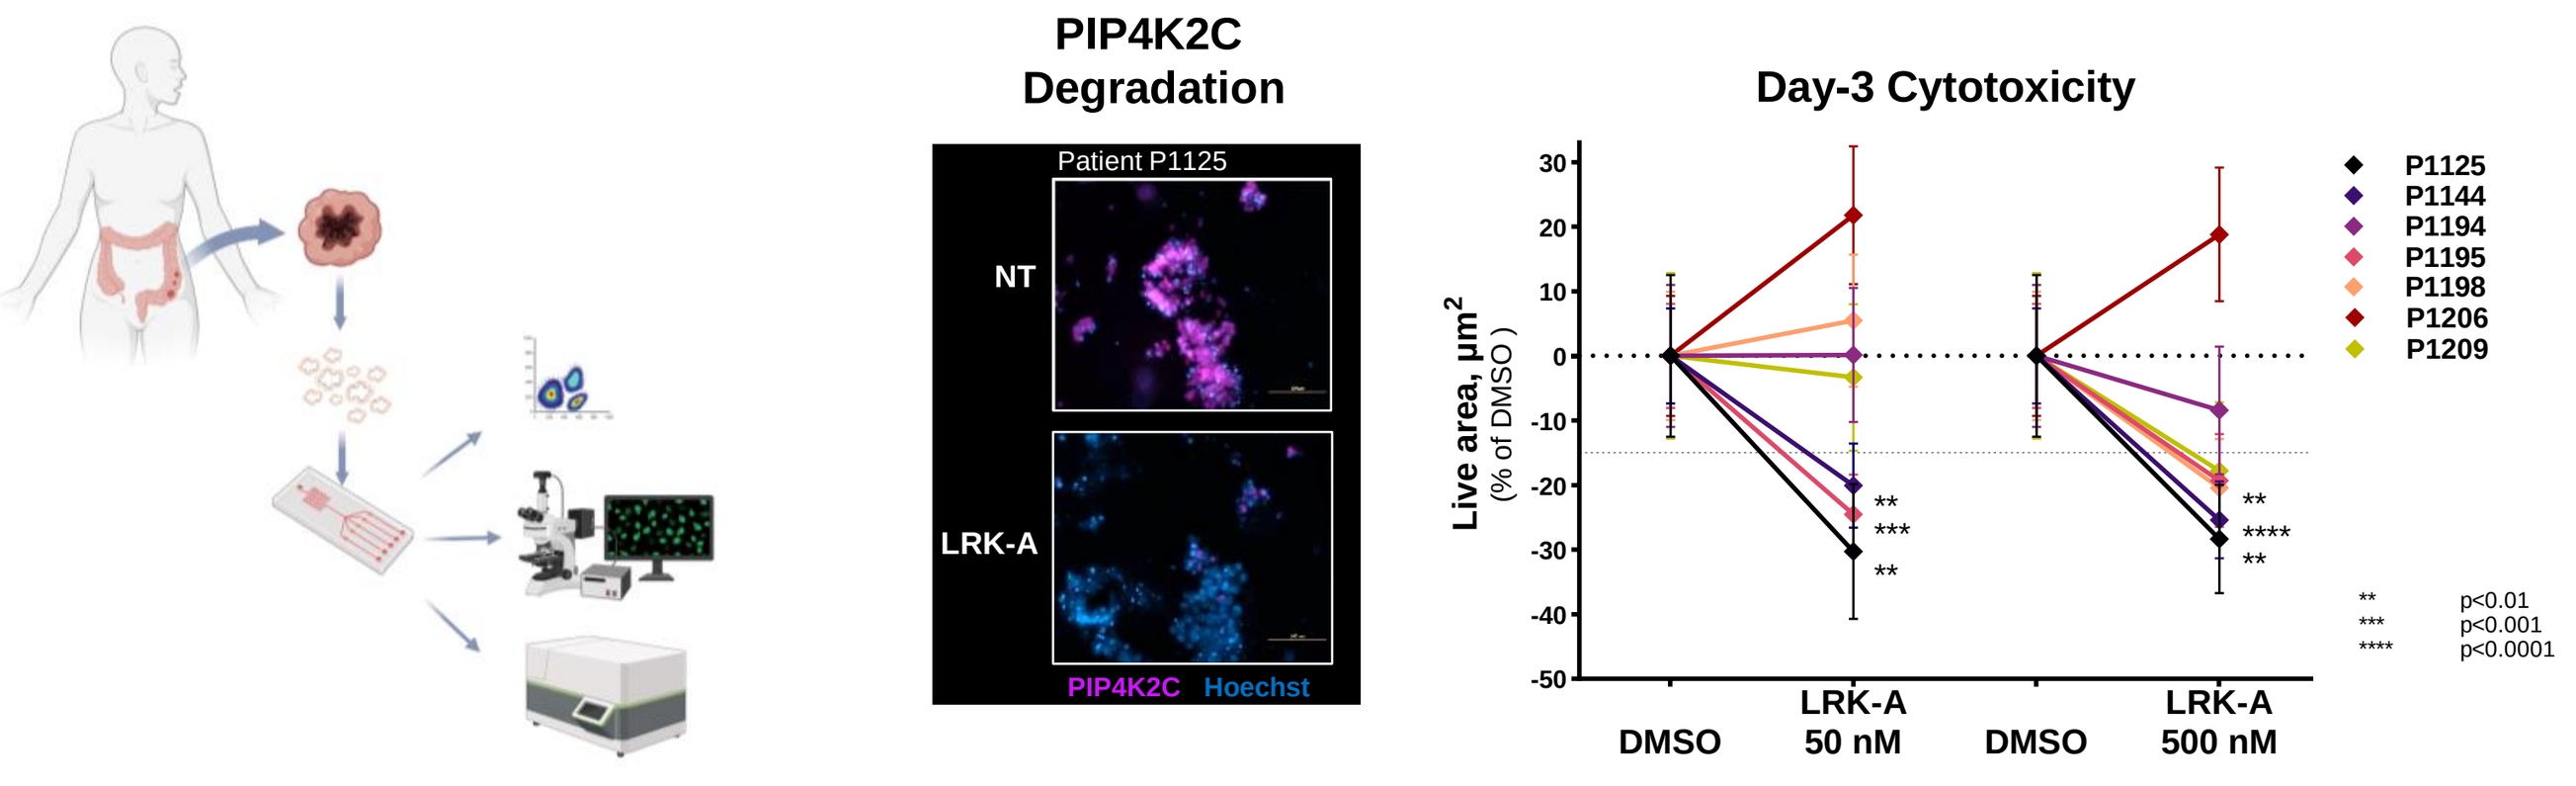
PIP4K2C Degradation
Patient P1125
NT
LRK-A
PIP4K2C Hoechst
Day-3 Cytotoxicity
Live area, µm² (% of DMSO)
P1125
P1144
P1194
P1195
P1198
P1206
P1209
** p<0.01
*** p<0.001
**** p<0.0001
DMSO
LRK-A 50 nM
DMSO
LRK-A 500 nM

Primary resections from 7 individual CRC patients were processed into spheroids, seeded into a set of microfluidic channels as indicated in schematic on the left (Biorender) and treated with DMSO or LRK-A at indicated concentrations for 5 days. (center) Degradation of PIP4K2C was verified in each sample by Immunofluorescence; representative patient sample is shown. (right) Cytotoxicity (%Live area) in LRK-A-treated samples compared to DMSO at day 3 is shown. Comparable results are seen at day 5 (data not shown).

FIGURE 4b – Pleiotropic effects of LRK-A on tumor immunogenicity, antigen uptake, and immune activation and cytotoxicity

Nanostring analyses of samples post-treatment (day 3 and 5) reveal numerous candidate biomarkers of mechanism and response captured over time. Pathways of immunogenic cell death and both myeloid and adaptive type-1 immunity (cytotoxic) and antigen presentation were enhanced at either day 3 or day 5 of culture in both patients analyzed to date. Collectively these preliminary findings validate the mechanistic hypothesis for PIP4K2C, and they illustrate how targeting PIP4K2C enhances anti-tumor immunity and counters cancer evasion through conjugated impacts in cancer cells and multiple immune cell subsets.

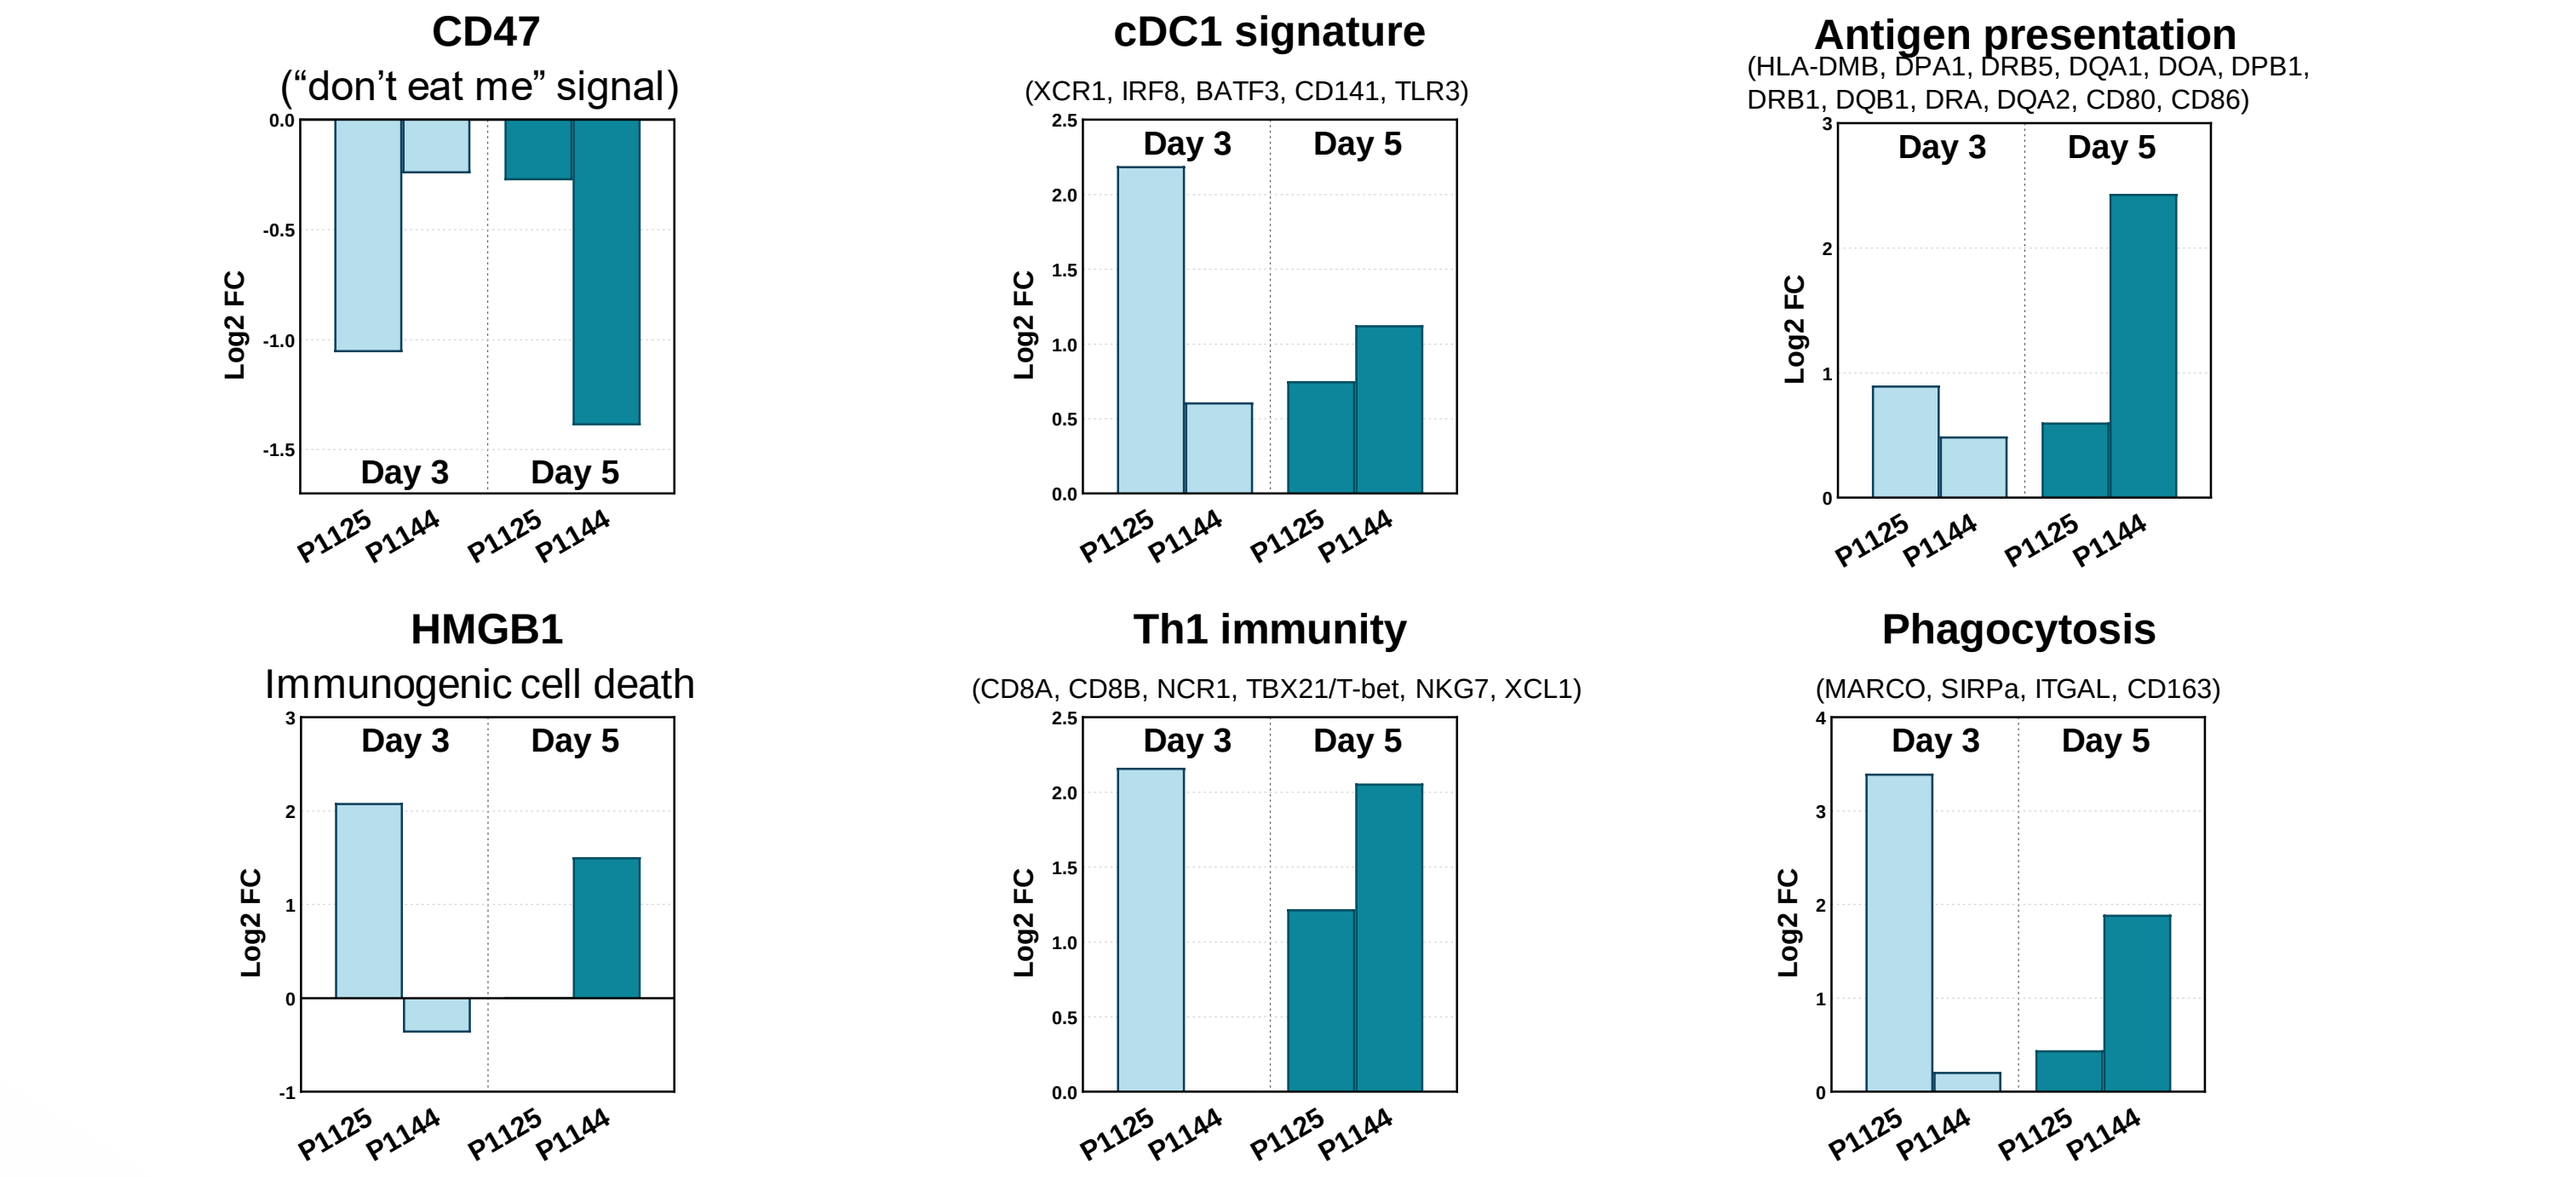
CD47
("don't eat me" signal)
cDC1 signature
(XCR1, IRF8, BATF3, CD141, TLR3)
Antigen presentation
(HLA-DMB, DPA1, DRB5, DQA1, DOA, DPB1, DRB1, DQB1, DRA, DQA2, CD80, CD86)
HMGB1
Immunogenic cell death
Th1 immunity
(CD8A, CD8B, NCR1, TBX21/T-bet, NKG7, XCL1)
Phagocytosis
(MARCO, SIRPa, ITGAL, CD163)
Log2 FC
Day 3
Day 5
P1125
P1144

Spheroid cultures were analyzed by Nanostring (IO panel) at days 3 and 5 of culture. Preliminary data available from two first patients at 500 nM are shown.

CONCLUSIONS AND PERSPECTIVES

- Collectively, our results show that PIP4K2C can be specifically and effectively targeted in vivo using a bifunctional degrader.
- The reduced tumor growth we observed upon targeting PIP4K2C alone suggests that PIP4K2C is a highly promising target for therapeutic intervention.
- Proof-of-concept efficacy in human patient samples ex vivo highlights the unique therapeutic potential of LRK-A in CRC.
- LRK-A development is ongoing with clinical studies anticipated in late 2024/early 2025.

Acknowledgments - We are grateful to the physicians, patients and their families who have consented to participate in this study. We also thank the consultants and CRO teams who have supported this work including The Bioinformatics CRO, Certara, Charles River Labs, MedGenome, ICE Biosciences, and Wuxi AppTec.

References

1. Wang DG, Paddock MN, Lundquist MR, et al. PIP4Ks suppress insulin signaling through a catalytic-independent mechanism. *Cell Rep*. 2019
2. Shim H, Wu C, Ramsamooj S, et al. Deletion of the gene PIP4K2C, a novel phosphatidylinositol kinase, results in hyperactivation of the immune system. *Proc Natl Acad Sci U S A*. 2016